Economic Burden of Cytomegalovirus Infection in CMV-Seropositive (CMV R+) Hematopoietic Stem Cell Transplant Recipients Managed With Pre-Emptive Therapy: A Single Center Experience

Memorial Sloan Kettering Cancer Center

Jiaqi Fang, MD, MPH[1], Phaedon D. Zavras, MD[1], Yiqi Su, MS[1], Amit D. Raval[2], PhD, Yuexin Tang[2], PhD, Miguel-Angel Perales, MD[2,3], Sergio A. Giralt, MD[2,3], Genovefa A. Papanicolaou, MD[1,3]

[1]Memorial Sloan Kettering Cancer Center, Infectious Disease Service, New York, NY, United States, [2]Merck & Co., Inc., Kenilworth, NJ, USA, [3]Memorial Sloan Kettering Cancer Center, Bone Marrow Transplant Service, New York, NY, United States, [4]Weill Cornell Medical College, Cornell University, New York, NY, United States

Abstract No: 521

Background

- Cytomegalovirus (CMV) infection is the most common viral infection in allogeneic hematopoietic stem cell transplantation (HCT) recipients
- Preemptive therapy (PET) is effective in preventing CMV end-organ disease (EOD) but poses substantial healthcare resource use (HCRU) and cost
- Established benchmarks for HCRU and cost in the era of PET are required to perform cost benefit analyses of novel interventions for CMV

Objectives

- To compare direct all-cause HCRU and cost in CMV R^+ by receipt of PET (PET versus no PET) through 180 days post HCT (D180)
- To quantify CMV-related HCRU and cost among PET recipients through D180
- To compare CMV-related with non CMV related HCRU and cost through D180

Methods

- **Study Design**: Retrospective cohort study
- **Study Population:** CMV R+ adult allogeneic HCT recipients of first peripheral blood or marrow allograft at a single center from 3/2013 to 12/2017
- **Data Sources:** Clinical data including PET reasons and dates of hospitalizations were extracted from the electronic medical records. Inpatient hospital charges were obtained from the Vizient billing database and converted to costs using institutional cost-to-charge ratios, wage index and inflation rate to 2017 US Dollars
- **CMV monitoring:** CMV+ recipients were monitored weekly by quantitative PCR assay starting on day 14 through D180 and managed by PET per standards of care
- **CMV risk: Patients were categorized in two mutually exclusive groups:** Low risk (LR) recipients of unmodified graft from matched related or unrelated donors or High risk (HR): recipients of unmodified graft from mismatched donor or ex vivo T-cell depleted graft from any donor.
- **PET group**: Receipt of pre-emptive antiviral therapy for CMV viremia
- **Follow-up period: Date of Index HCT** to the earliest of D180 or death/relapse/2nd transplant, whichever occurred first.
- **Healthcare resource utilization:**
 - Length of inpatient stay (LOS) for index admission, and subsequent readmissions by day 180 post HCT
 - Number of all-cause and CMV-related readmissions by 180 post HCT
 - CMV-related readmissions were defined as readmissions for initiation of PET, work up or management of CMV End Organ Disease and any readmission where PET was initiated

Baseline Characteristics

- Of 368 HCT recipients, 192 (52%) were HR and 176 (48%) LR
- Overall, 208 (56.5%) patients received PET
- HR comprised 72% of PET group but only 26% of No PET group.

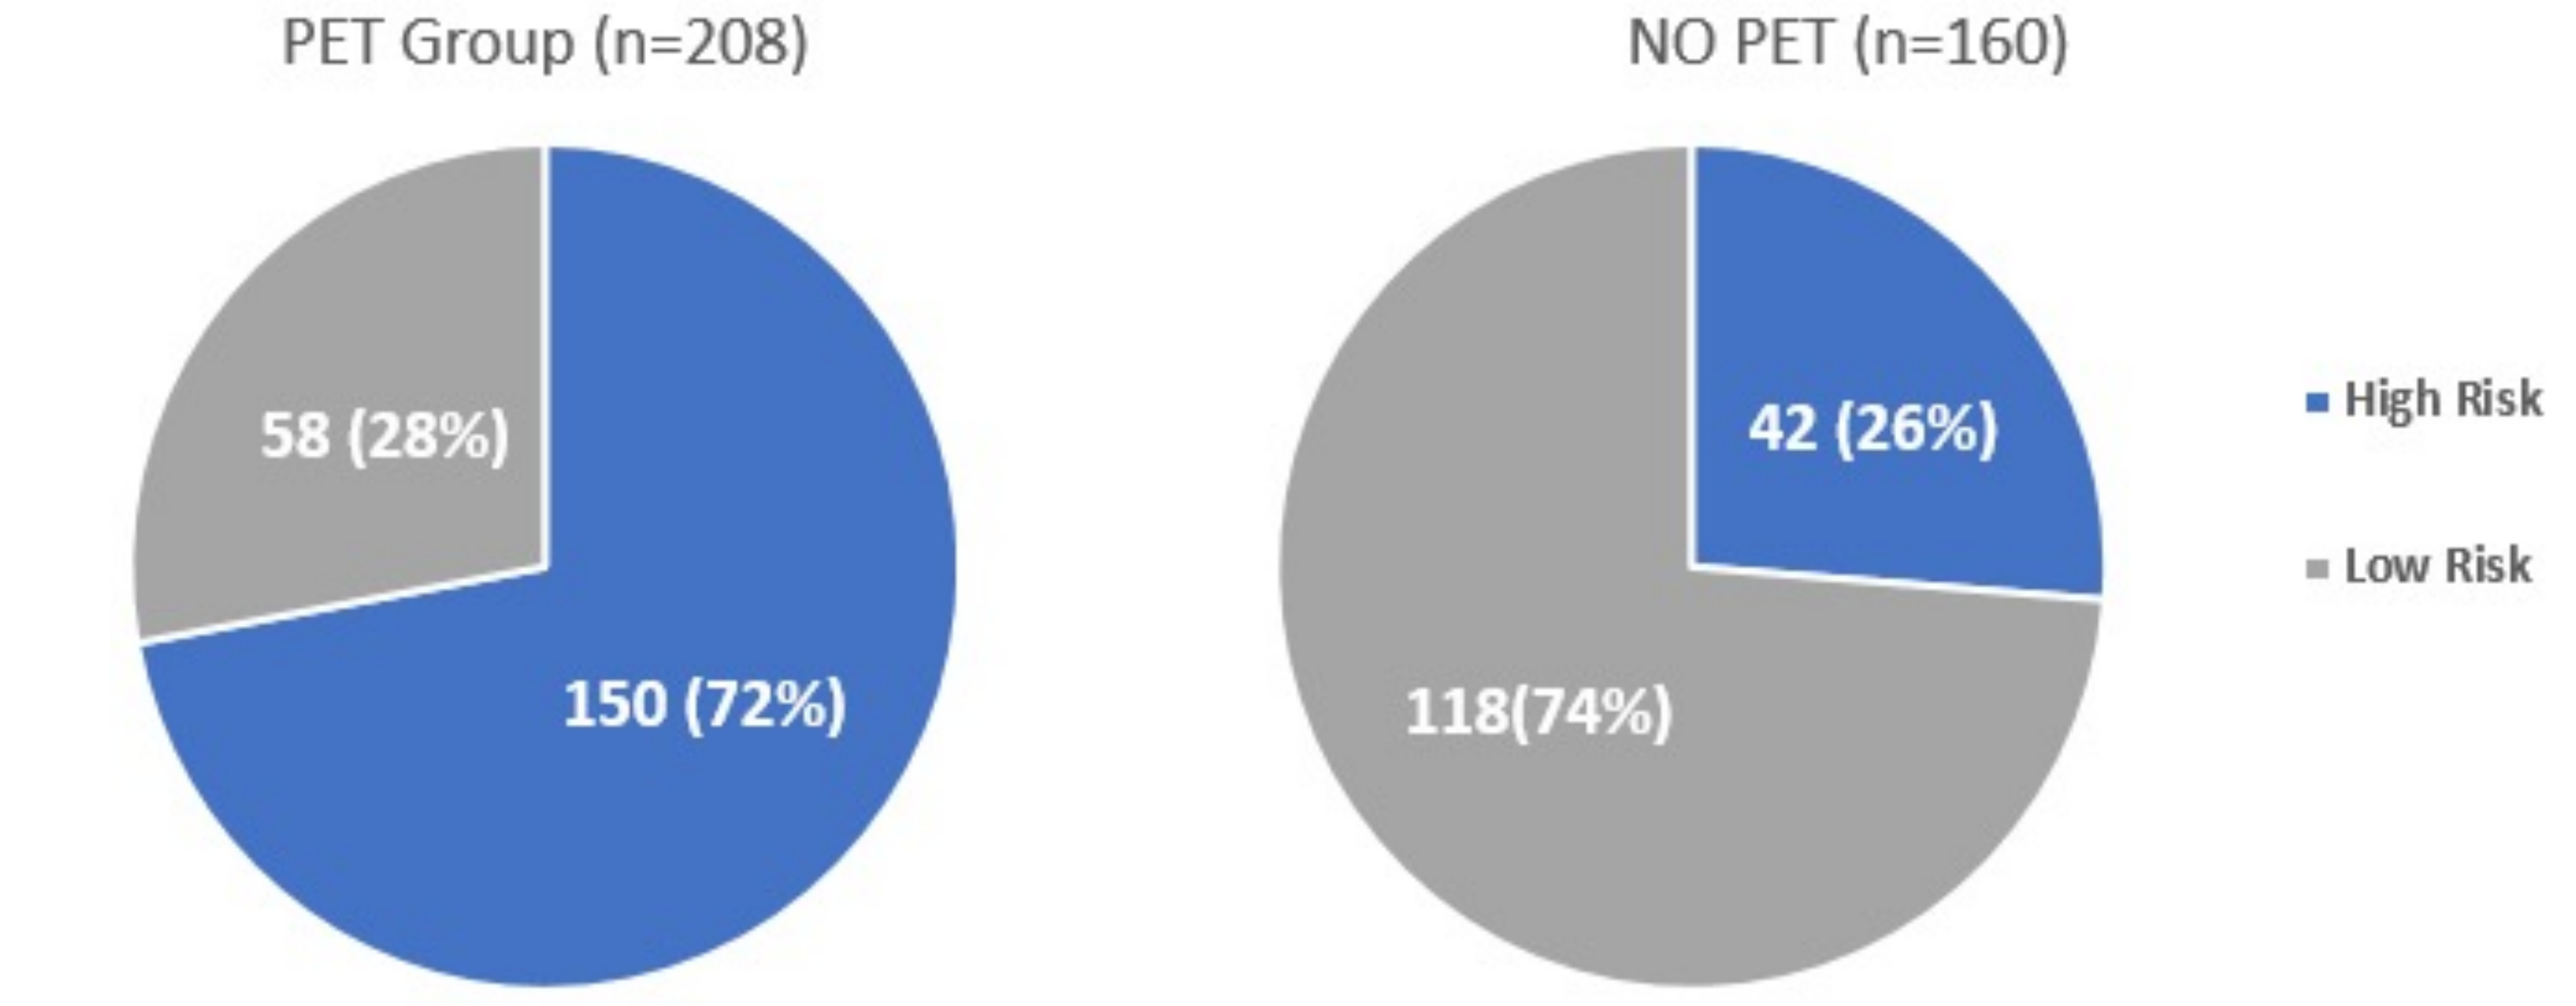
PET Group (n=208)
NO PET (n=160)
58 (28%)
150 (72%)
42 (26%)
118(74%)
High Risk
Low Risk

Results

Comparison of average LOS (days) by PET use

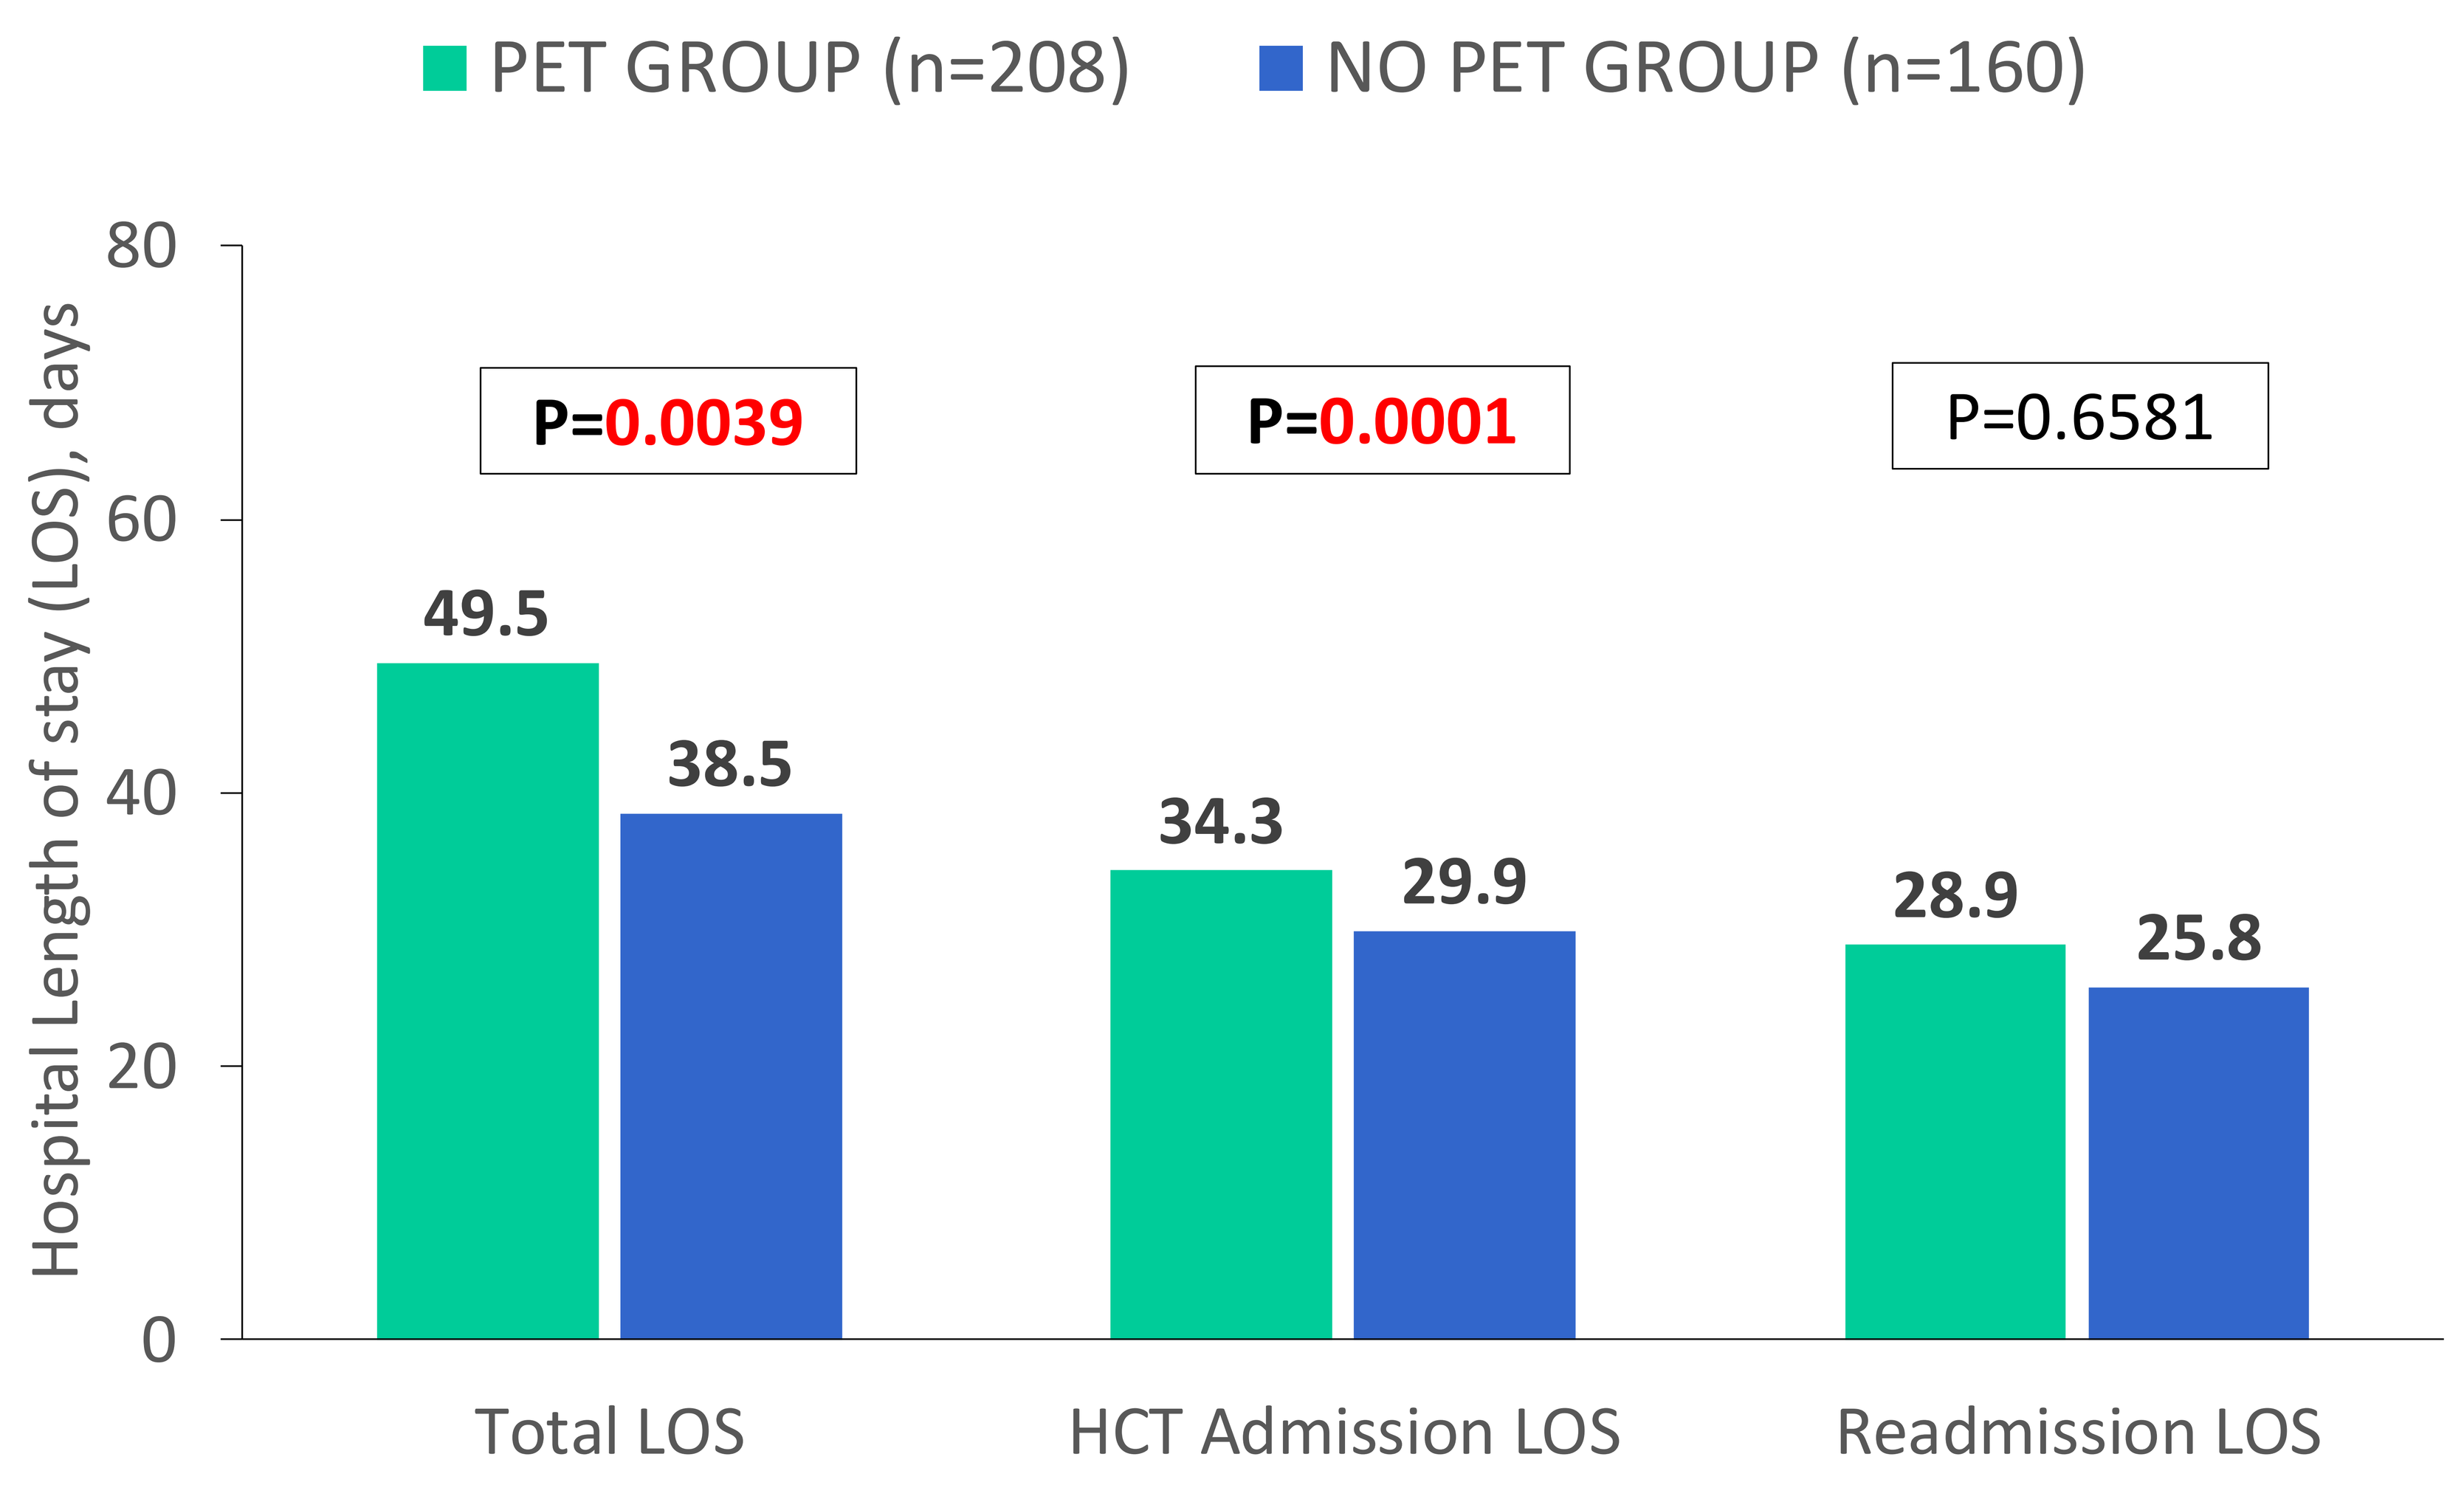
PET GROUP (n=208)
NO PET GROUP (n=160)
Hospital Length of stay (LOS), days
0
20
40
60
80
P=0.0039
P=0.0001
P=0.6581
49.5
38.5
34.3
29.9
28.9
25.8
Total LOS
HCT Admission LOS
Readmission LOS

PET group had longer total LOS from D0 through D180 (p=**0.0039**) and longer LOS for index HCT admission (p=**0.0001**) compared with NO PET group

Comparison of average cost per patient by PET use

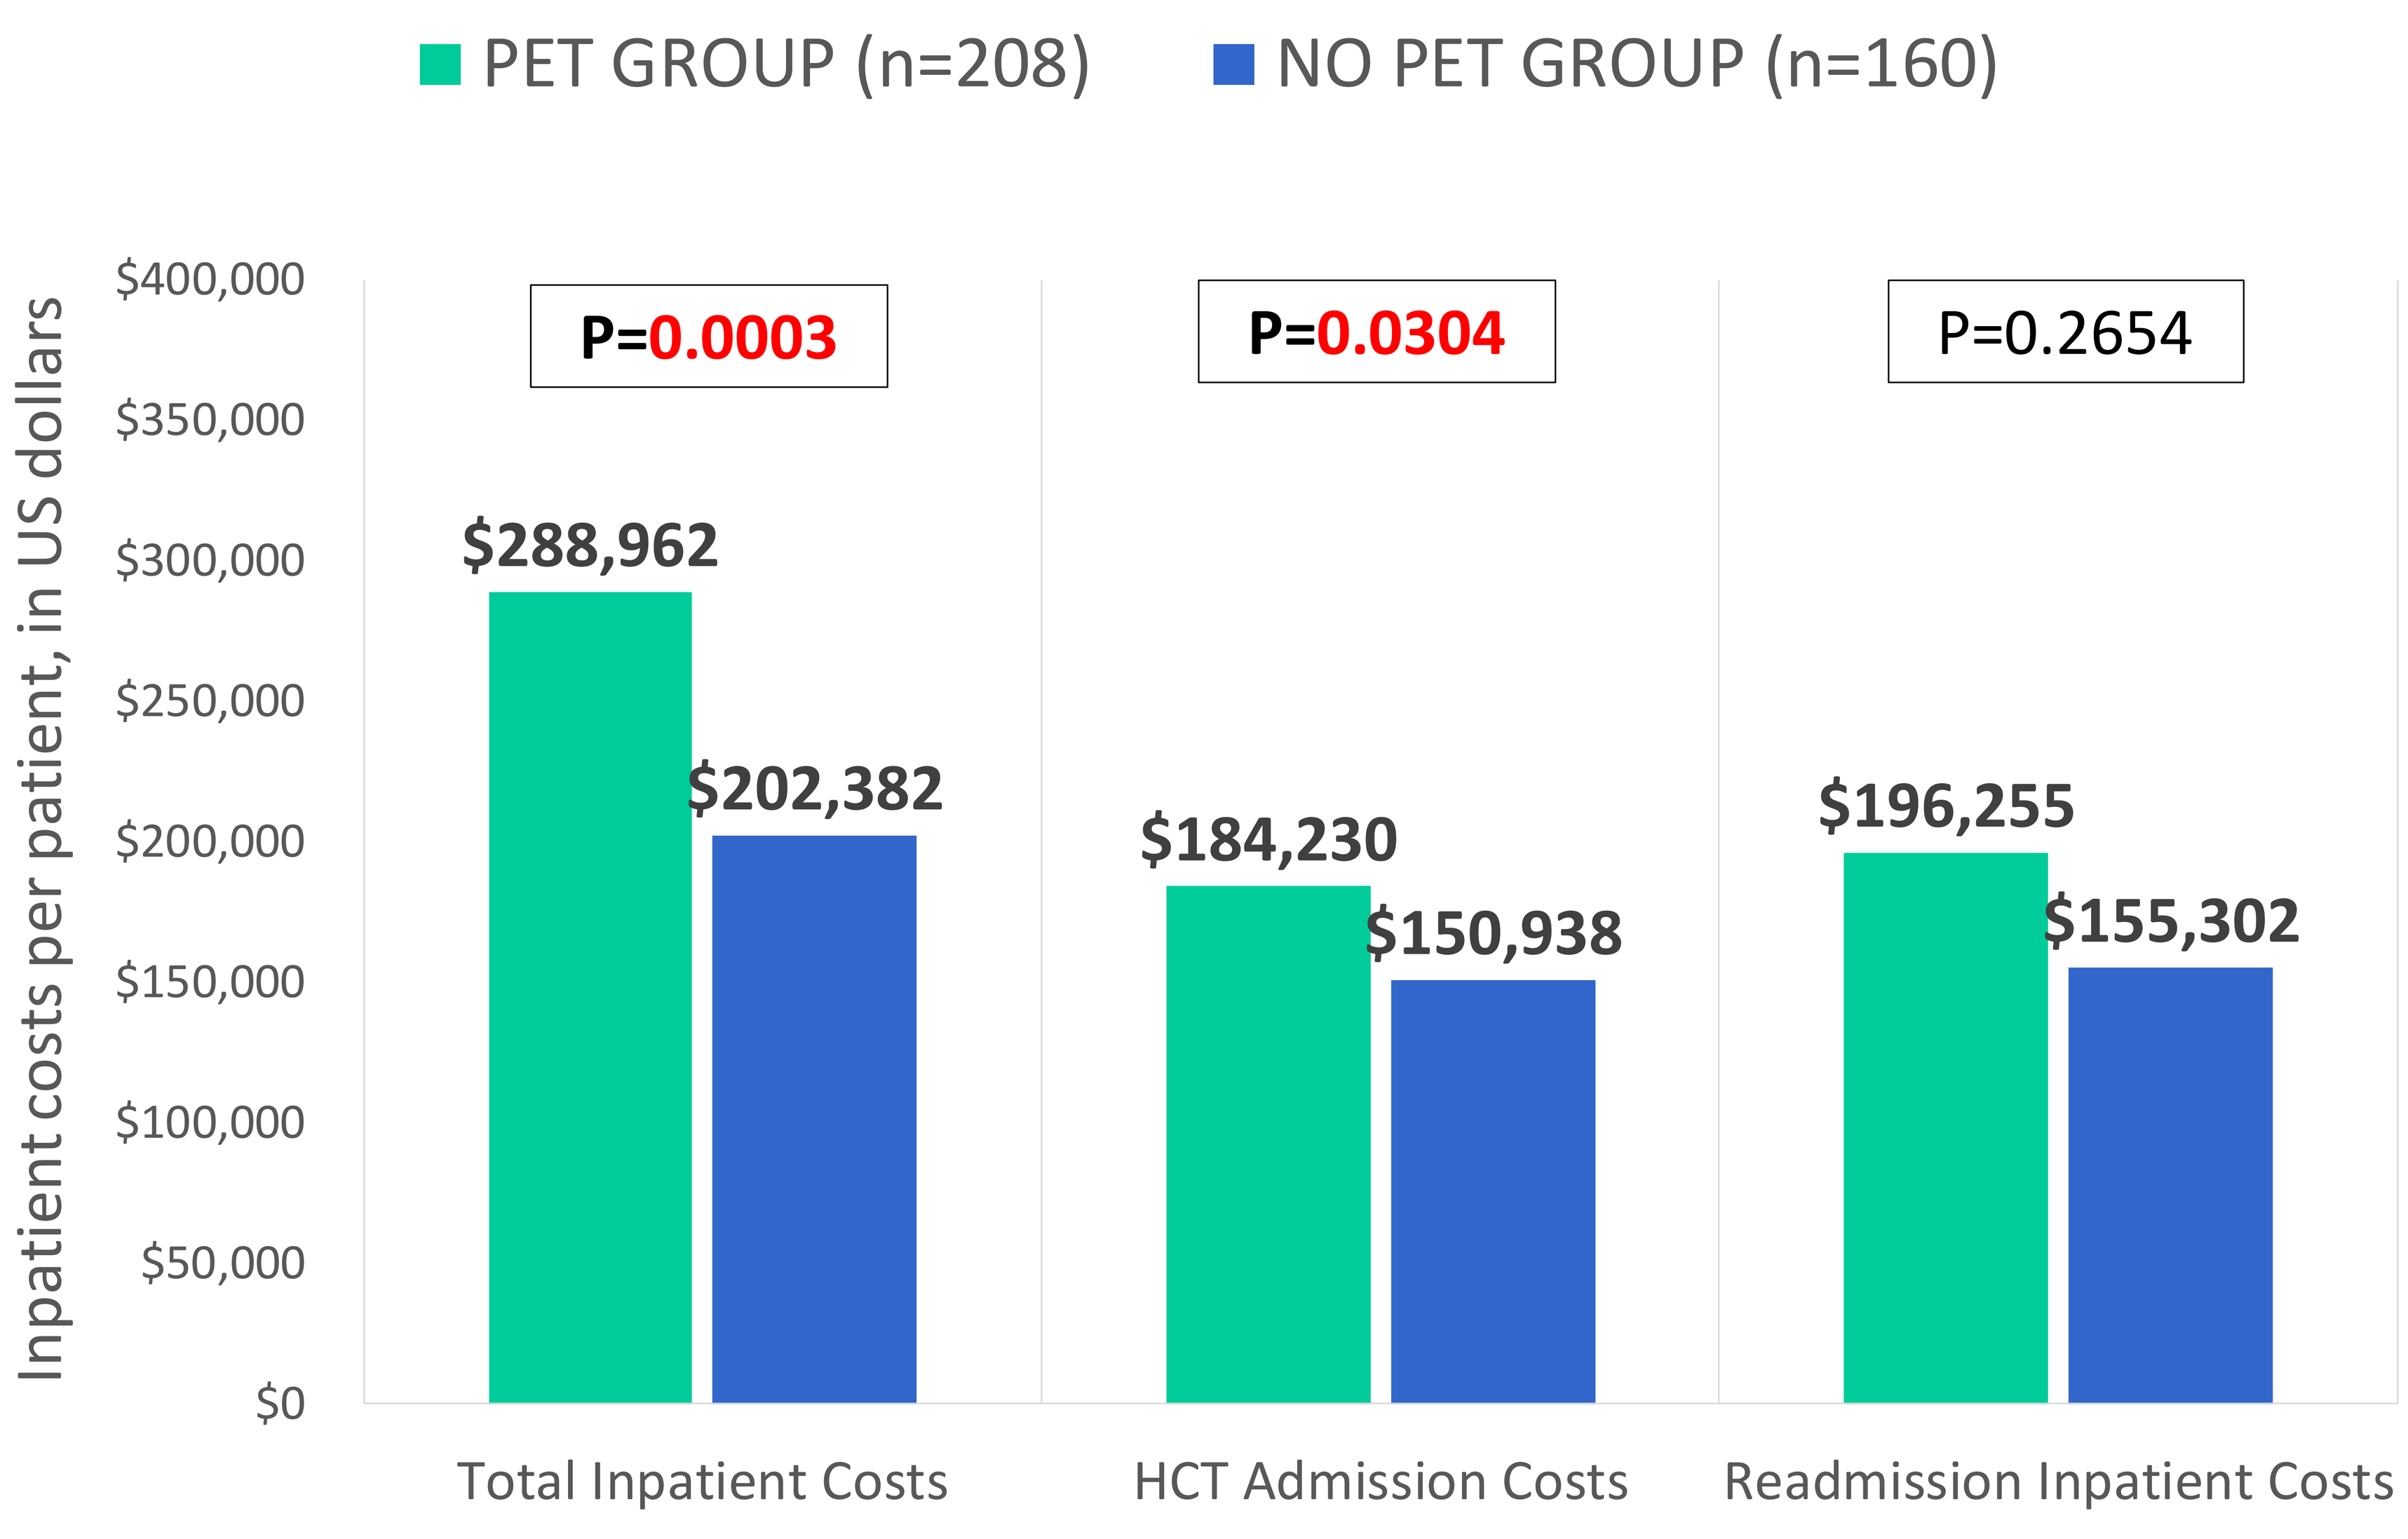
PET GROUP (n=208)
NO PET GROUP (n=160)
Inpatient costs per patient, in US dollars
$0
$50,000
$100,000
$150,000
$200,000
$250,000
$300,000
$350,000
$400,000
P=0.0003
P=0.0304
P=0.2654
$288,962
$202,382
$184,230
$150,938
$196,255
$155,302
Total Inpatient Costs
HCT Admission Costs
Readmission Inpatient Costs

PET group had higher total average inpatient cost per patient through D180 (p=**0.0003**) and for index HCT admission (p=**0.0304**) compared with no PET group.

Proportion of CMV related admissions and costs

NUMBER (%) BY TYPES OF READMISSIONS

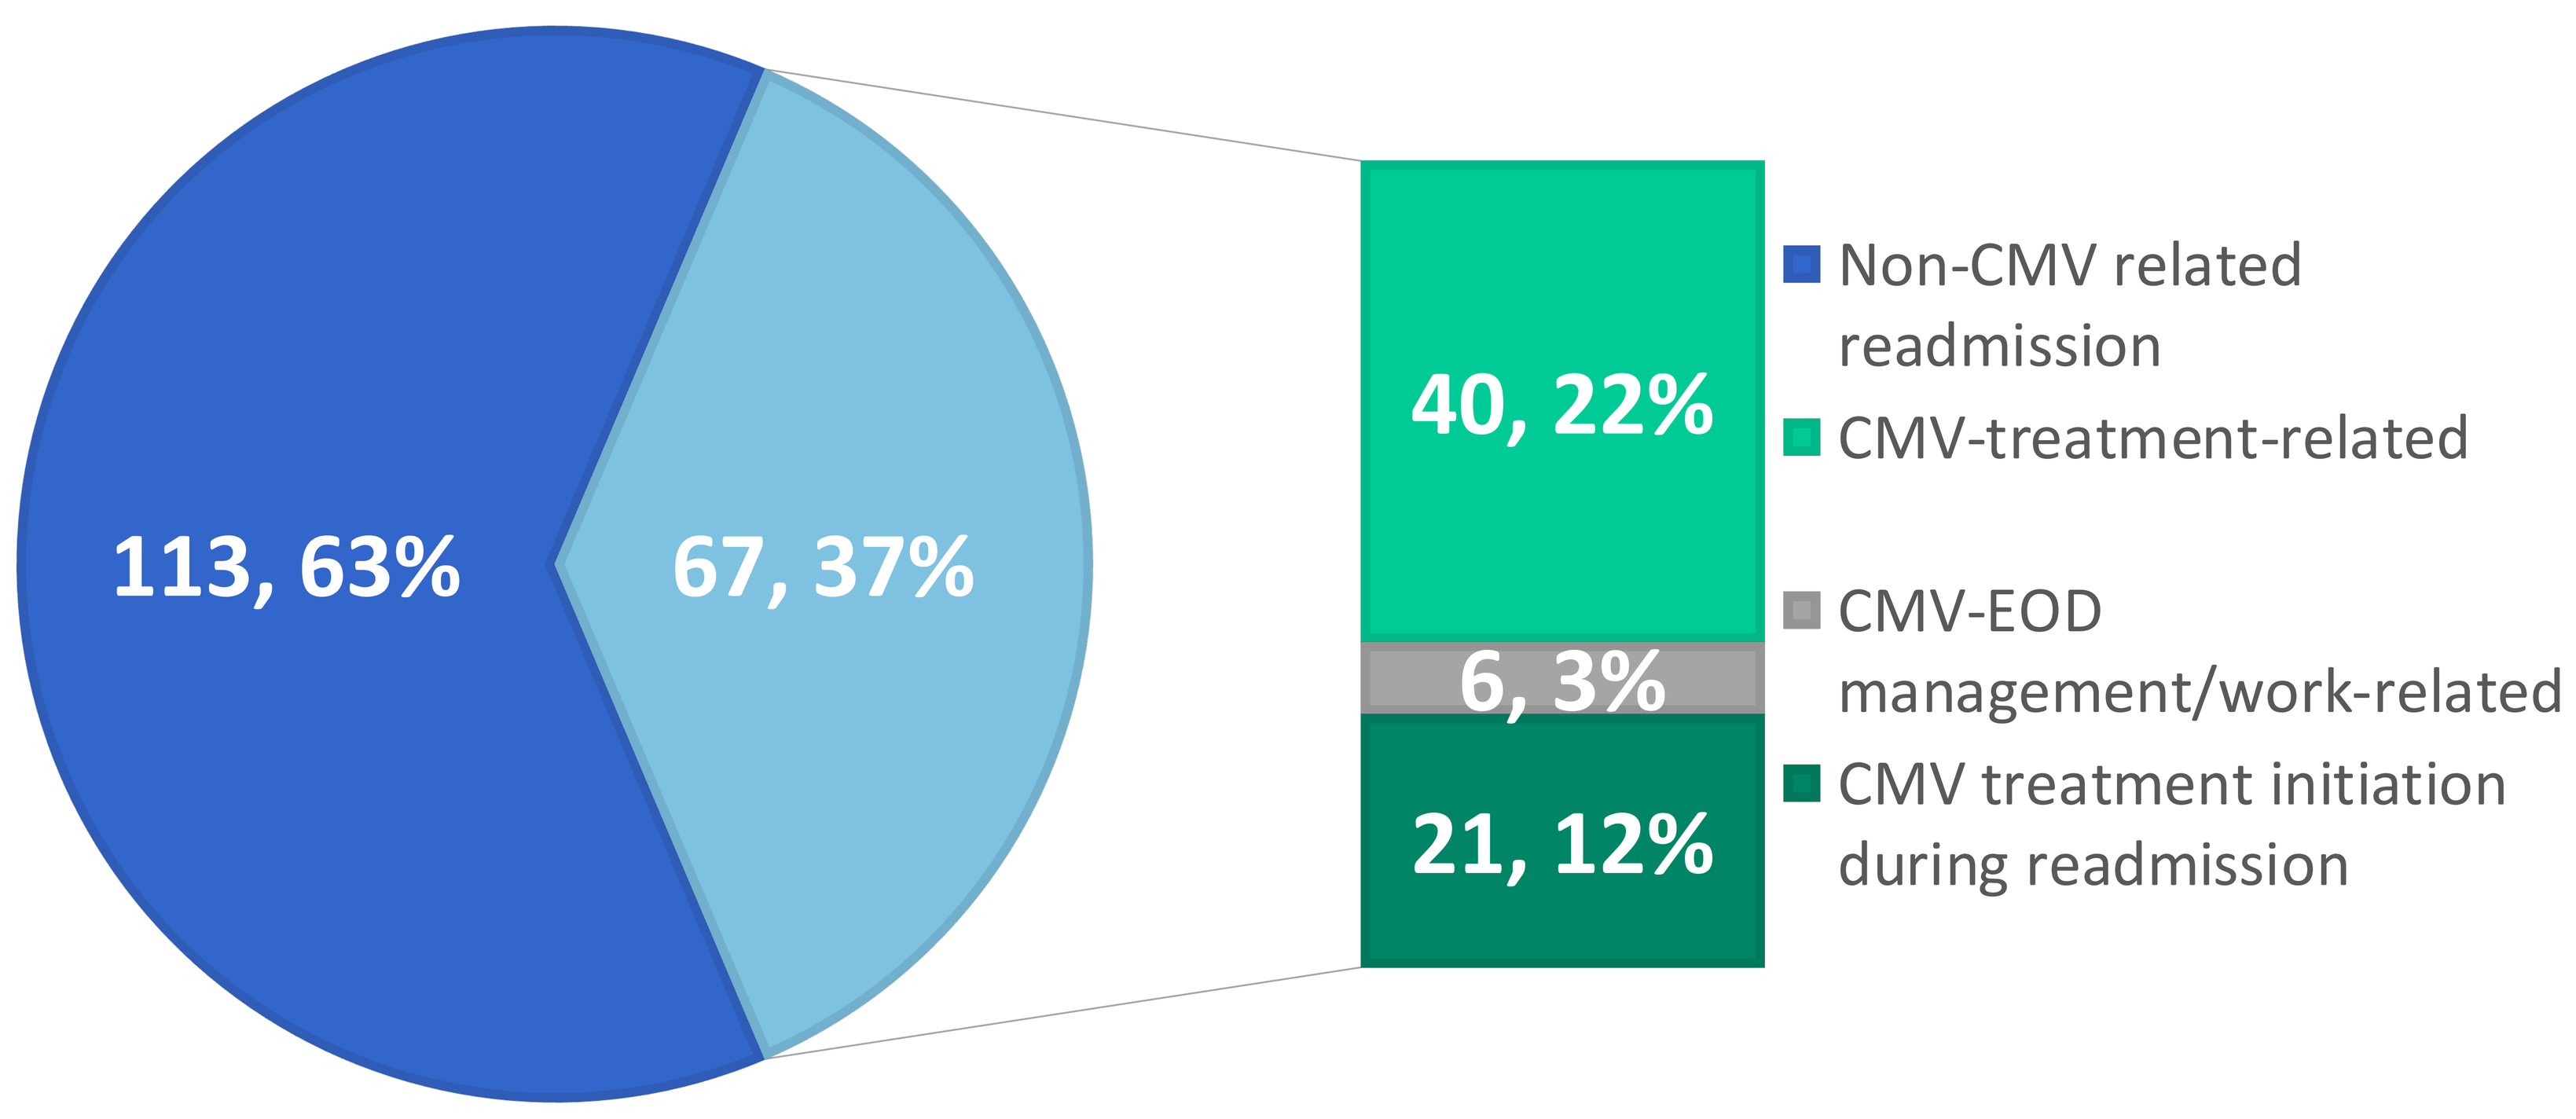
113, 63%
67, 37%
40, 22%
6, 3%
21, 12%
Non-CMV related readmission
CMV-treatment-related
CMV-EOD management/work-related
CMV treatment initiation during readmission

% OF TOTAL READMISSION COST SHARE BY TYPES OF READMISSION

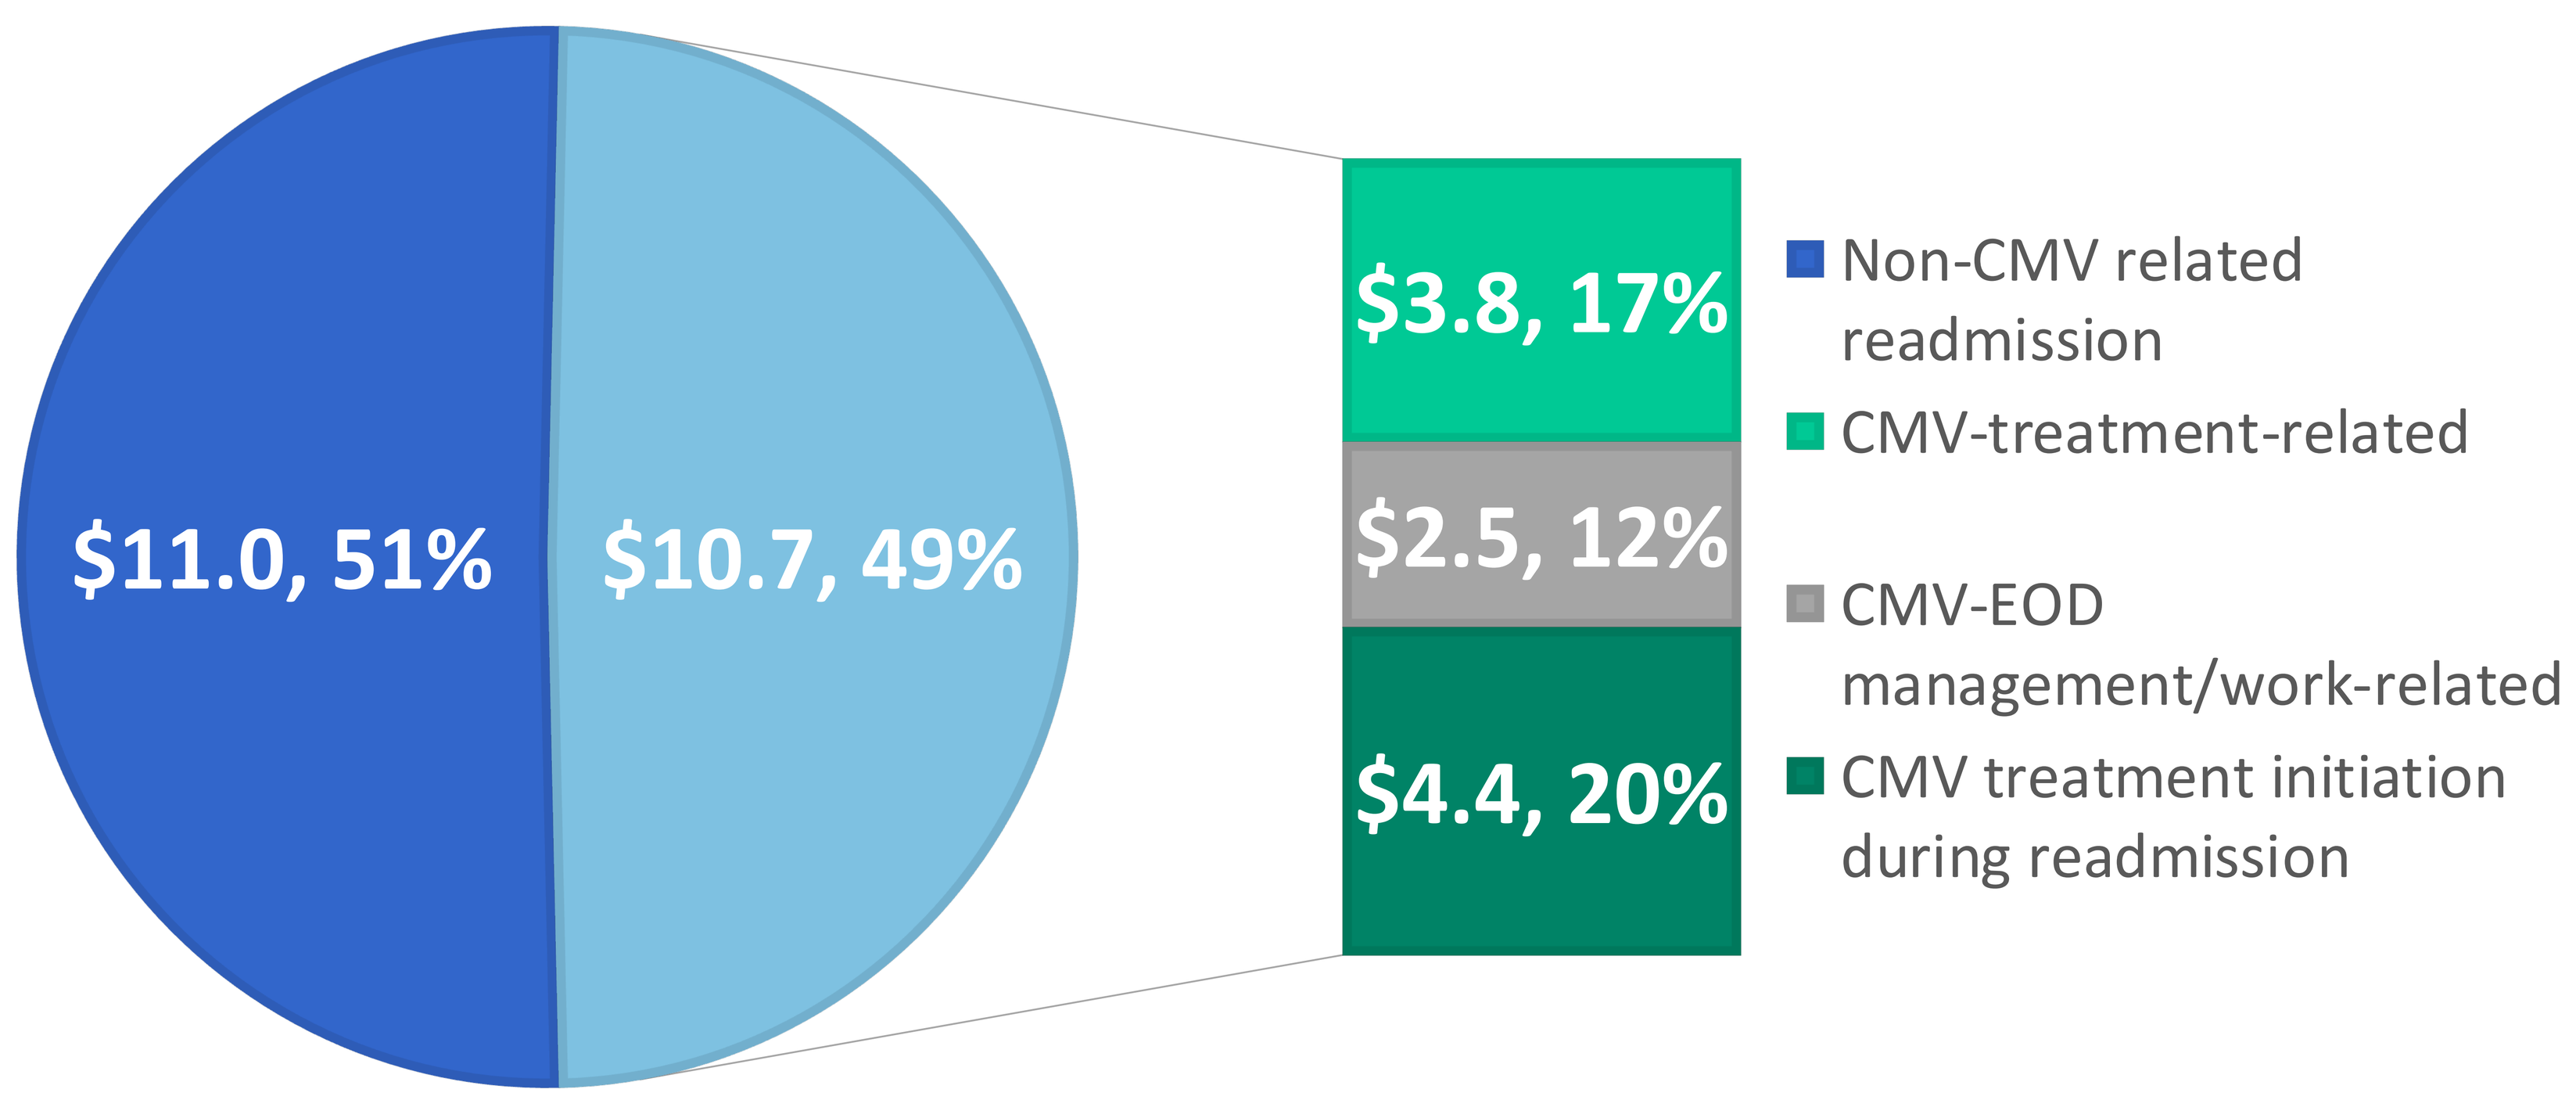
$11.0, 51%
$10.7, 49%
$3.8, 17%
$2.5, 12%
$4.4, 20%
Non-CMV related readmission
CMV-treatment-related
CMV-EOD management/work-related
CMV treatment initiation during readmission

- CMV-related readmissions had higher average cost per episode compared with non CMV-related readmissions in the PET group ($167,701 versus $96,941, p=**0.0089**)
- In the PET group, CMV-related readmissions comprised 37% of all readmissions and incurred 49% of total readmission costs through D180.

Conclusion

- PET recipients incurred higher total HCRU and cost through D180 as compared with no PET recipients
- Among the PET recipients, CMV-related readmissions had a disproportionately high share of total inpatient readmissions costs compared to Non-CMV related readmissions
- Future studies are needed to examine the cost-effectiveness of alternative strategies for CMV management

Disclosure: This study was funded by a grant from Merck & Co., Inc